Clinical trial simulation and hypothesis testing using amyloid pathology longitudinal translational model

ISB M Institute for Systems Biology Moscow

Tatiana Karelina[1*], Oleg Demin[1], Sridhar Duvvuri[2], Tim Nicholas[2]

1. Institute for Systems Biology, Moscow, Russia.
2. Pfizer Worldwide Research and Development, Groton, CT, USA

INTRODUCTION

QSP model describing amyloid secretion, distribution and aggregation was developed earlier. It successfully describes multiple of data on monkey, human, mice. It allows description of longitudinal amyloid progression. Here we apply it for simulation of clinical trial results and amyloid toxicity hypothesis testing.

METHODS

Amyloid pathology model

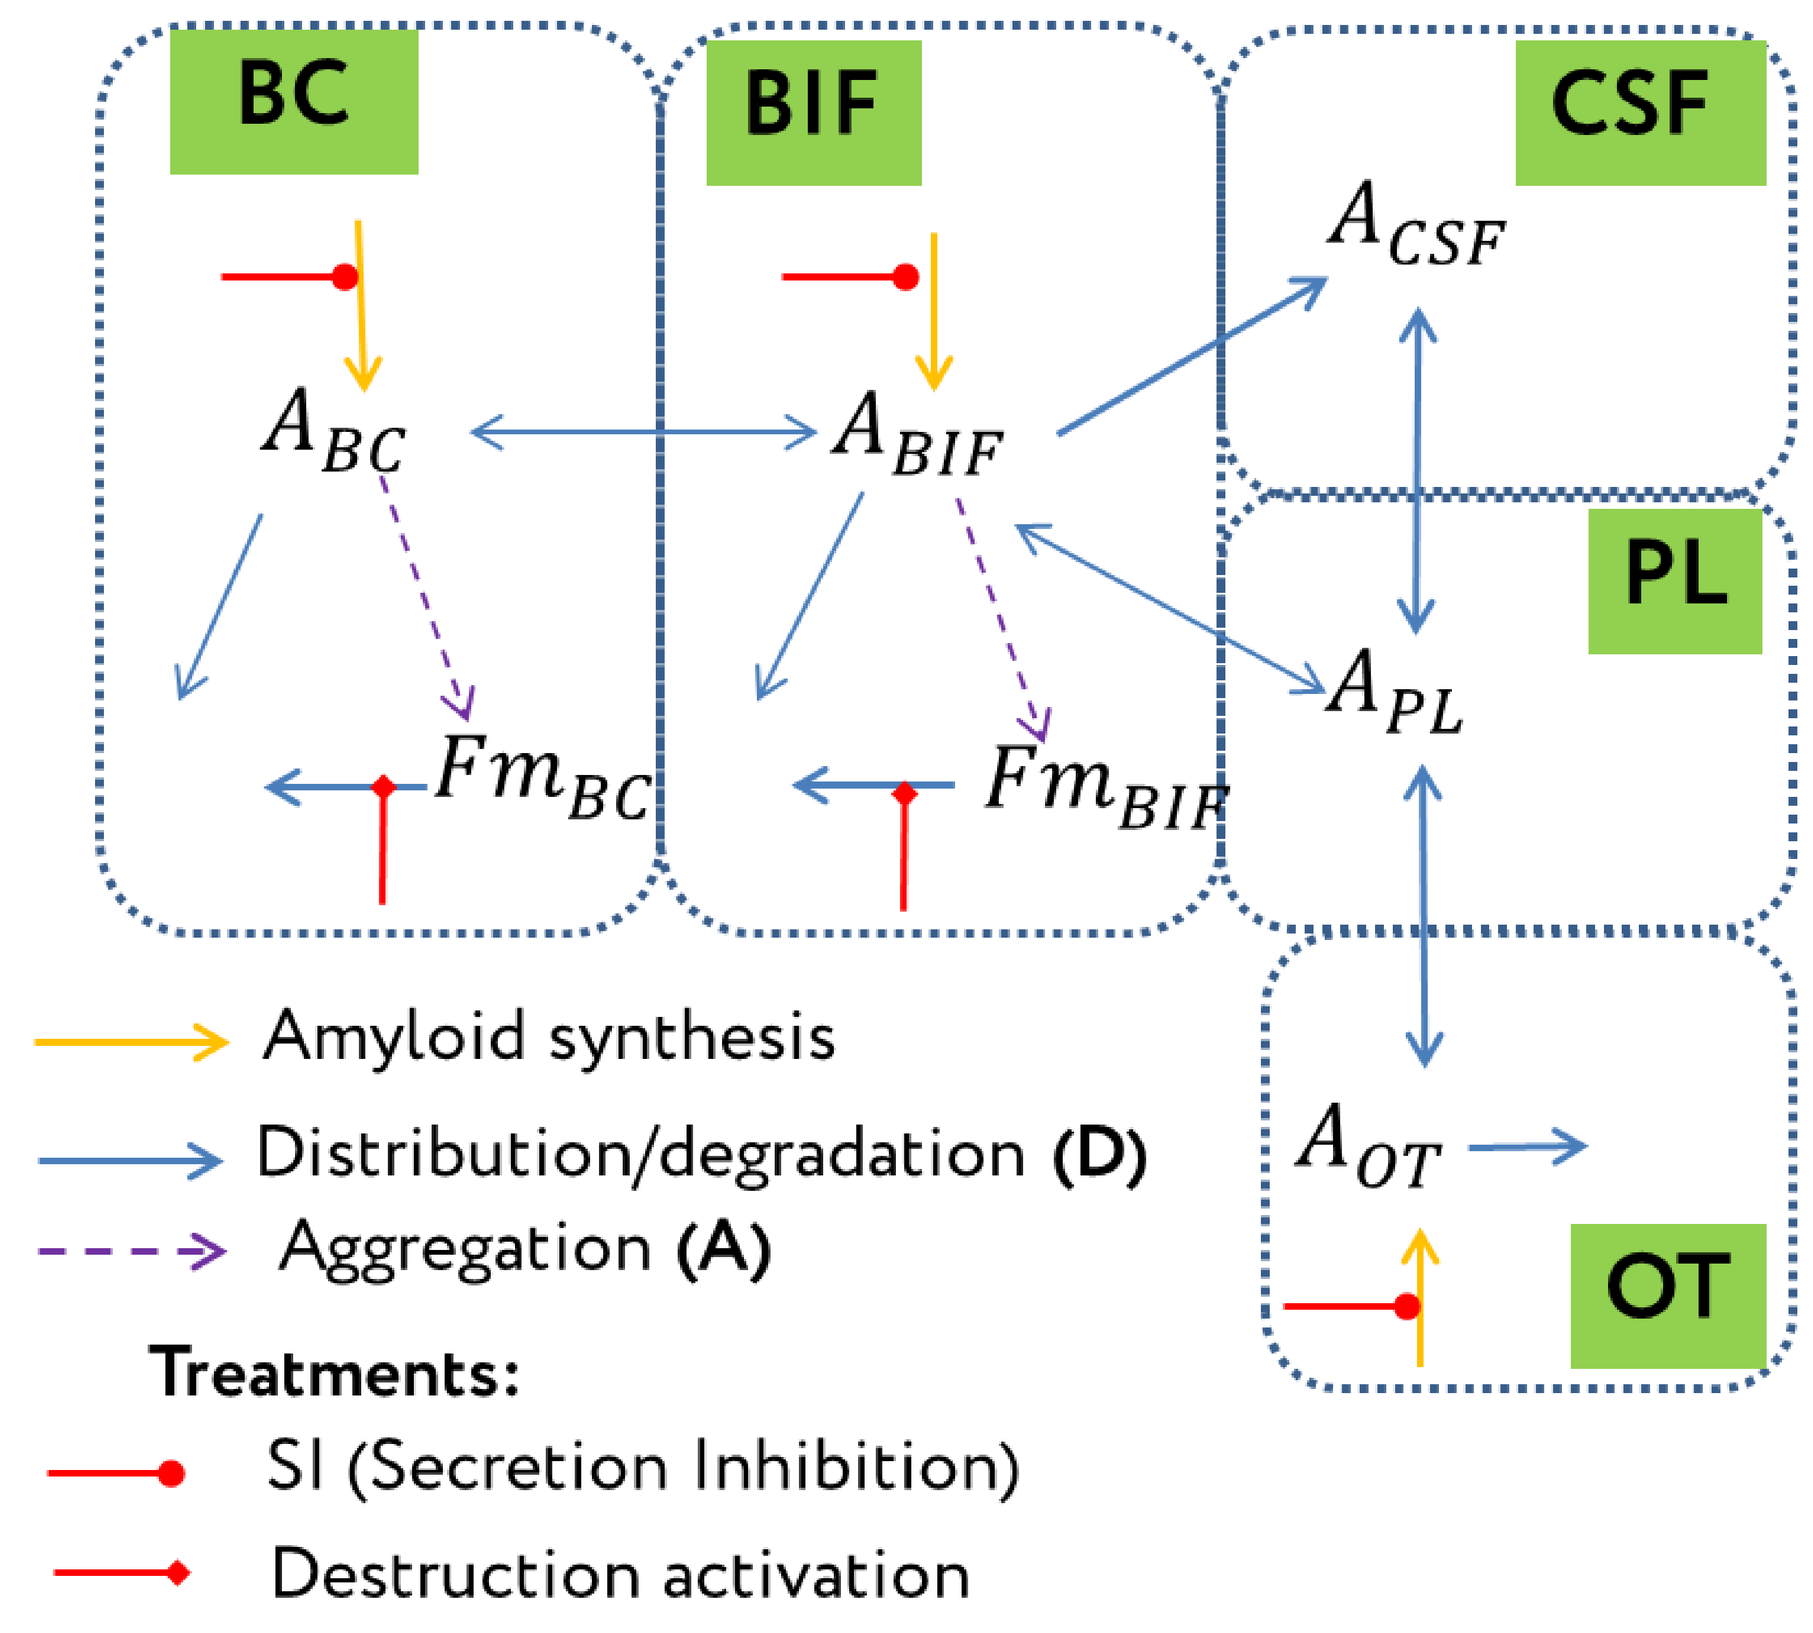
BC
BIF
CSF
PL
OT
A_{BC}
A_{BIF}
A_{CSF}
A_{PL}
A_{OT}
Fm_{BC}
Fm_{BIF}
Amyloid synthesis
Distribution/degradation (D)
Aggregation (A)
Treatments:
SI (Secretion Inhibition)
Destruction activation

Fig. 1. Scheme of model of Ab distribution and aggregation

Fm_{BIF} - insoluble Ab40(42)

Longitudinal progression model

- Progression during 20 years starting at ~ 60.
- Healthy and AD populations have the same baseline (and parameters) before 60 years.
- Healthy and AD populations may have different prog and Ai.

$$\mathrm{P}i = Pi_0 \cdot (1 + Ai \cdot Age_dep)$$

$$\text{Age_dep}=1-\frac{1}{1+Kin\cdot\exp((prog\cdot(t-t_{decline}))}$$

D-model: Ab time profiles in brain, CSF and plasma of 129/SVE mouse after administration of 125I-labeled Ab and oral administration of Avagacestat, steady-state values in CSF, PL, BR for human and mice, SILK data for human, Semagacestat PD data

A-model: Measured concentrations in BR, CSF, PL for AD, healthy at different ages, validation on PET data

Cognitive toxicity model

- Ab42 soluble was chosen as the key toxic specie;
- There are no irreversible changes in the system;
- Empirical choice of parameters for Adas-cog score: gradual change from 8 to approximately 35 (Rosenberg et al 2013) during overall disease progression.

Table 1. The list of hypotheses of Ab toxicity

Toxicity of amyloid	Ab is obligatory	#	Description
Proportional	No	H 1.1	$tox(Ab_{42}) = k_{dr} \cdot (Asoluble_{BR}^{42} - base_health)$, $base_health$=1 nM
	Yes	H 2.1	Same as 1.1, but takes into account **decrease of cognition** for the low Ab_{42} levels [Puzzo et al]
Threshold (prion type)	No	H 1.2	$tox(Ab_{42}) = k_{thr} * (t - t_{st})\ if\ Asoluble_{BR}^{42} > base_health$
	Yes	H 2.2	Same as 1.2, 2 but takes into account **decrease of cognition** for the low Ab_{42} levels
Proportional and threshold	No	H 1.3	combination of two influences: Ab is driver of disease, and after achieving threshold independent growth is added to Ab toxicity.

RESULTS

Amyloid pathology model performance

Fig. 2. Model calibration (examples)

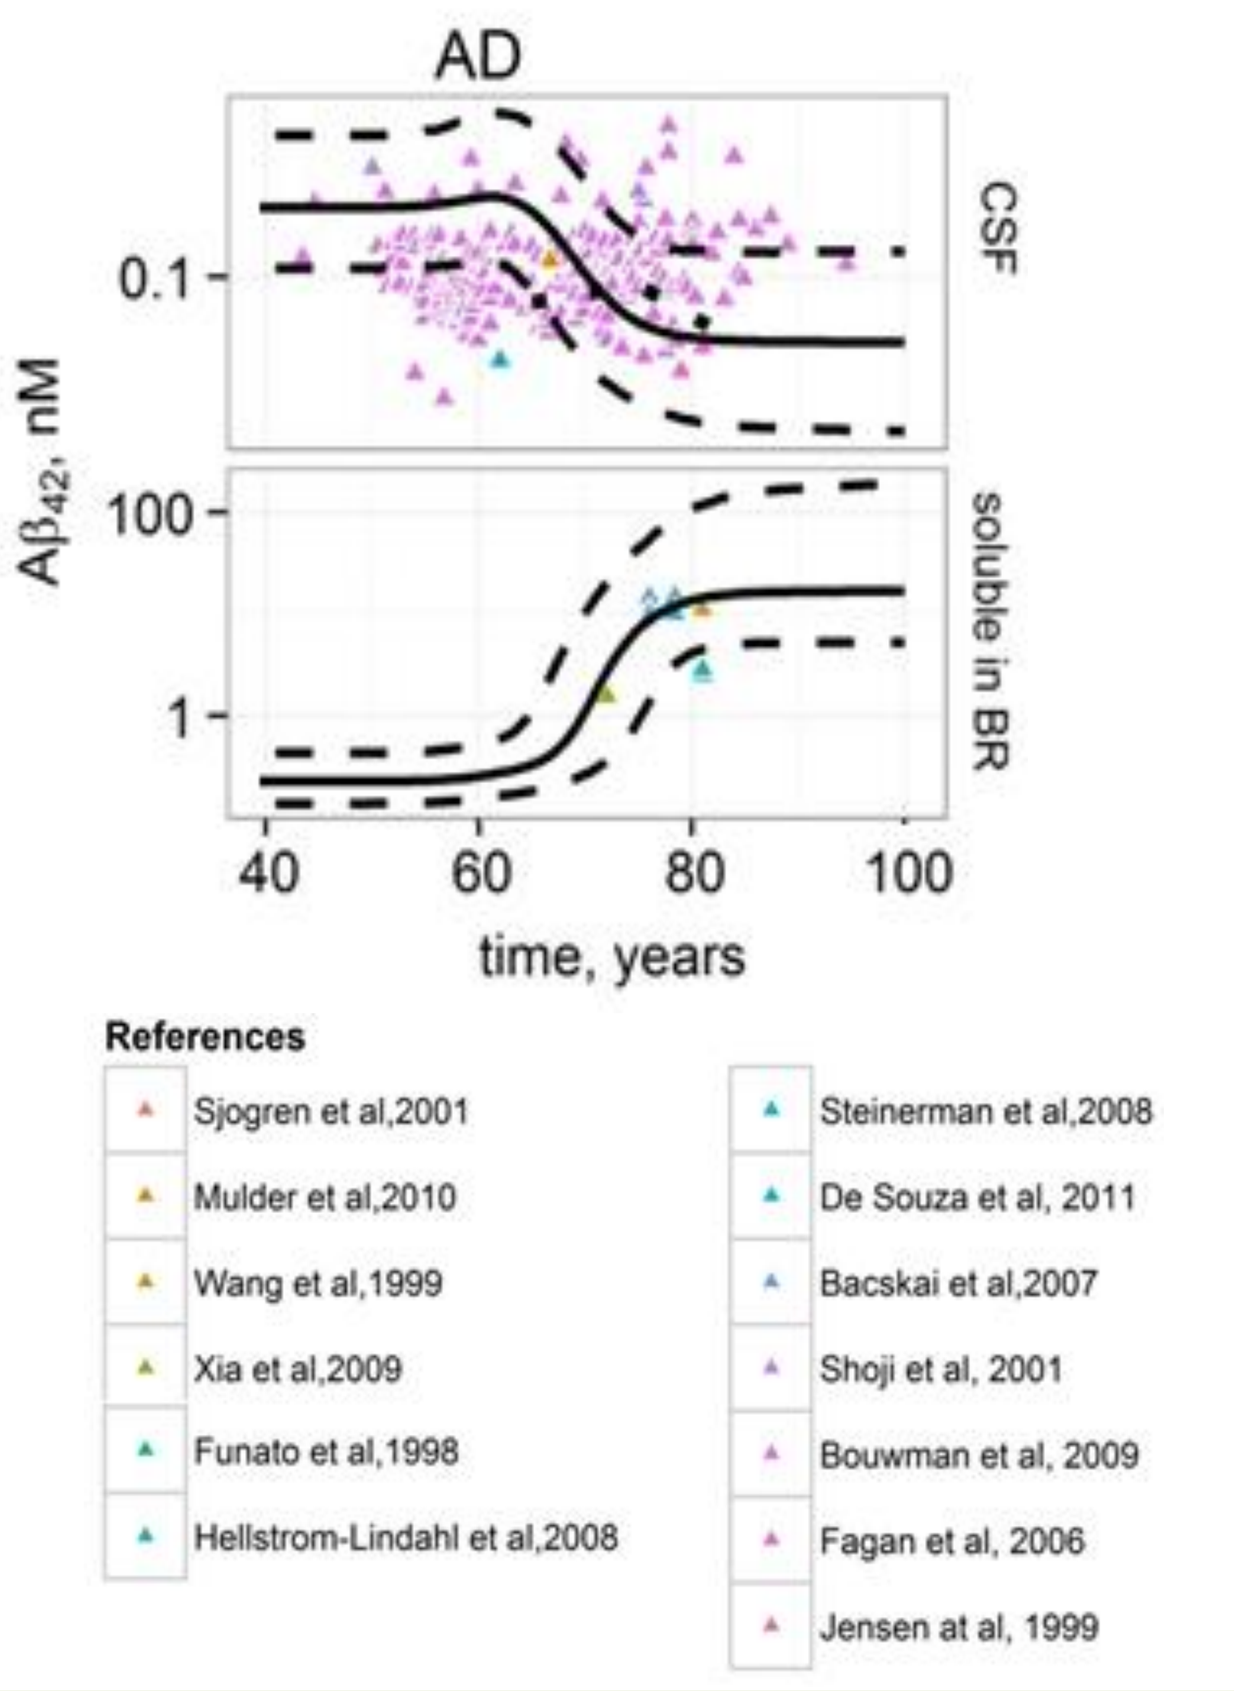
AD
CSF
soluble in BR
Aβ42, nM
time, years
References
Sjogren et al,2001
Mulder et al,2010
Wang et al,1999
Xia et al,2009
Funato et al,1998
Hellstrom-Lindahl et al,2008
Steinerman et al,2008
De Souza et al, 2011
Bacskai et al,2007
Shoji et al, 2001
Bouwman et al, 2009
Fagan et al, 2006
Jensen at al, 1999

Fig. 3. Model validation

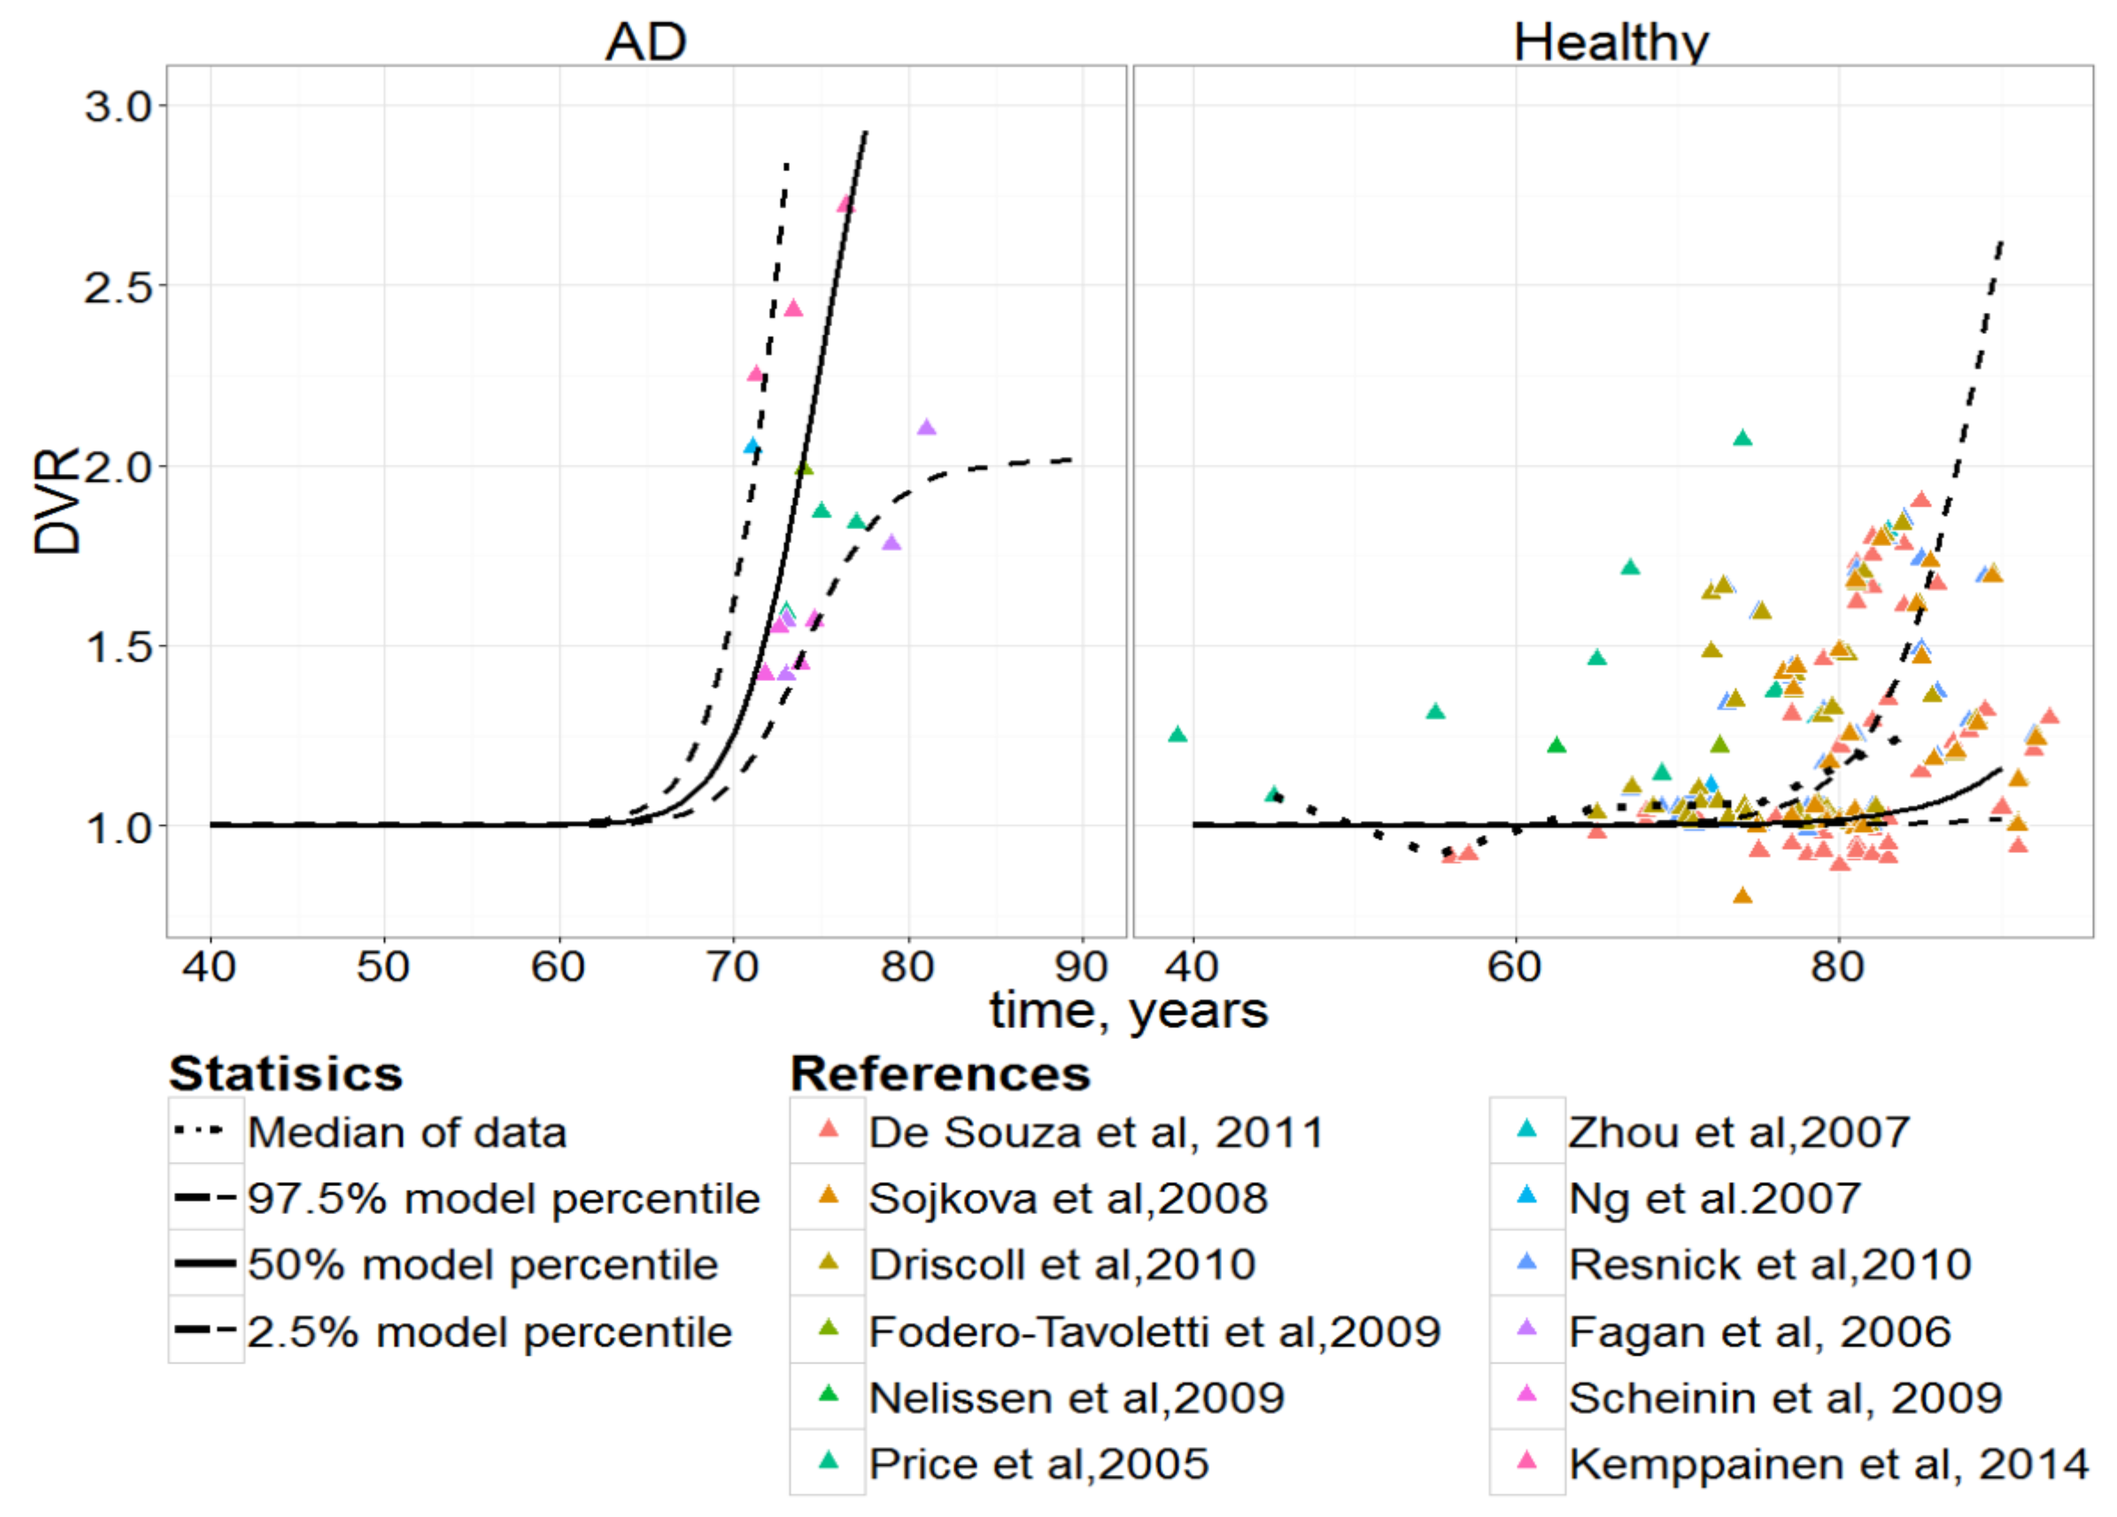
AD
Healthy
DVR
time, years
Statisics
Median of data
97.5% model percentile
50% model percentile
2.5% model percentile
References
De Souza et al, 2011
Sojkova et al,2008
Driscoll et al,2010
Fodero-Tavoletti et al,2009
Nelissen et al,2009
Price et al,2005
Zhou et al,2007
Ng et al.2007
Resnick et al,2010
Fagan et al, 2006
Scheinin et al, 2009
Kemppainen et al, 2014

Fig. 4. Simulations of Adas-cog dynamics under different hypotheses and different treatments

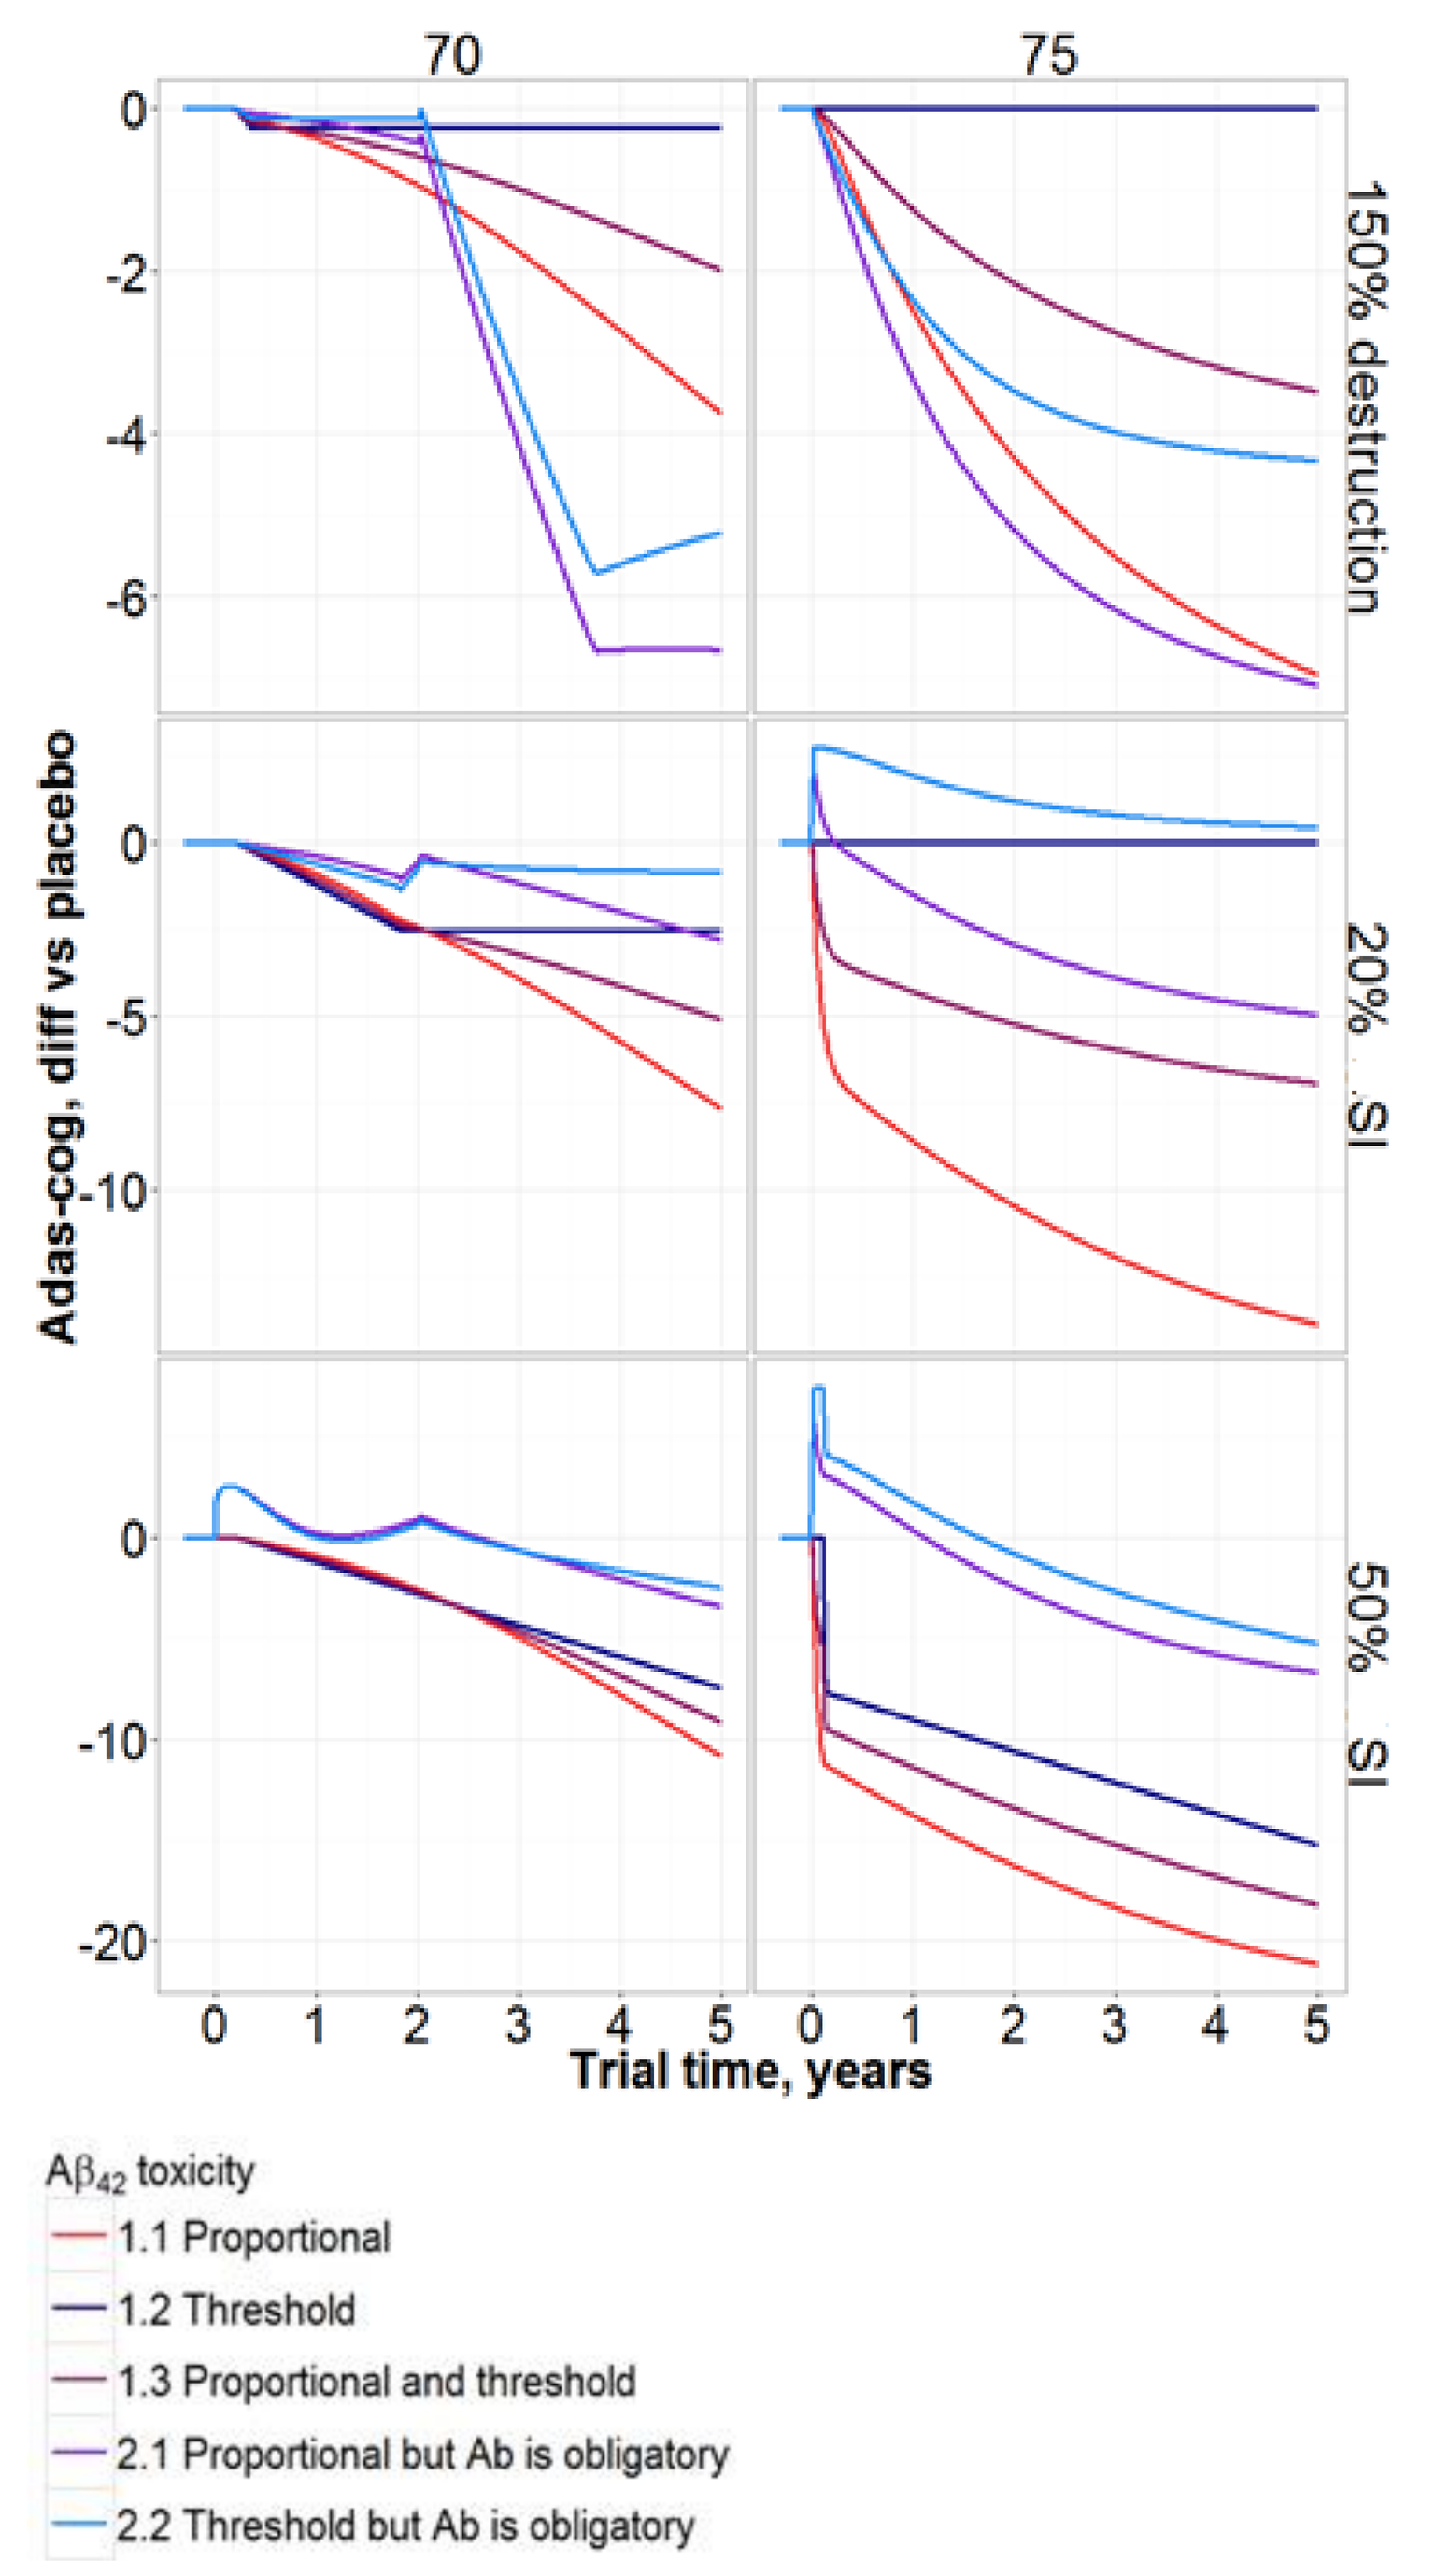
Age of start, years
70
75
150% destruction
20% SI
50% SI
Adas-cog, diff vs placebo
Trial time, years
Aβ42 toxicity
1.1 Proportional
1.2 Threshold
1.3 Proportional and threshold
2.1 Proportional but Ab is obligatory
2.2 Threshold but Ab is obligatory

Fig. 5. Simulations of Adas-cog, Ab42 in CSF and DVR under hypothesis 2.1

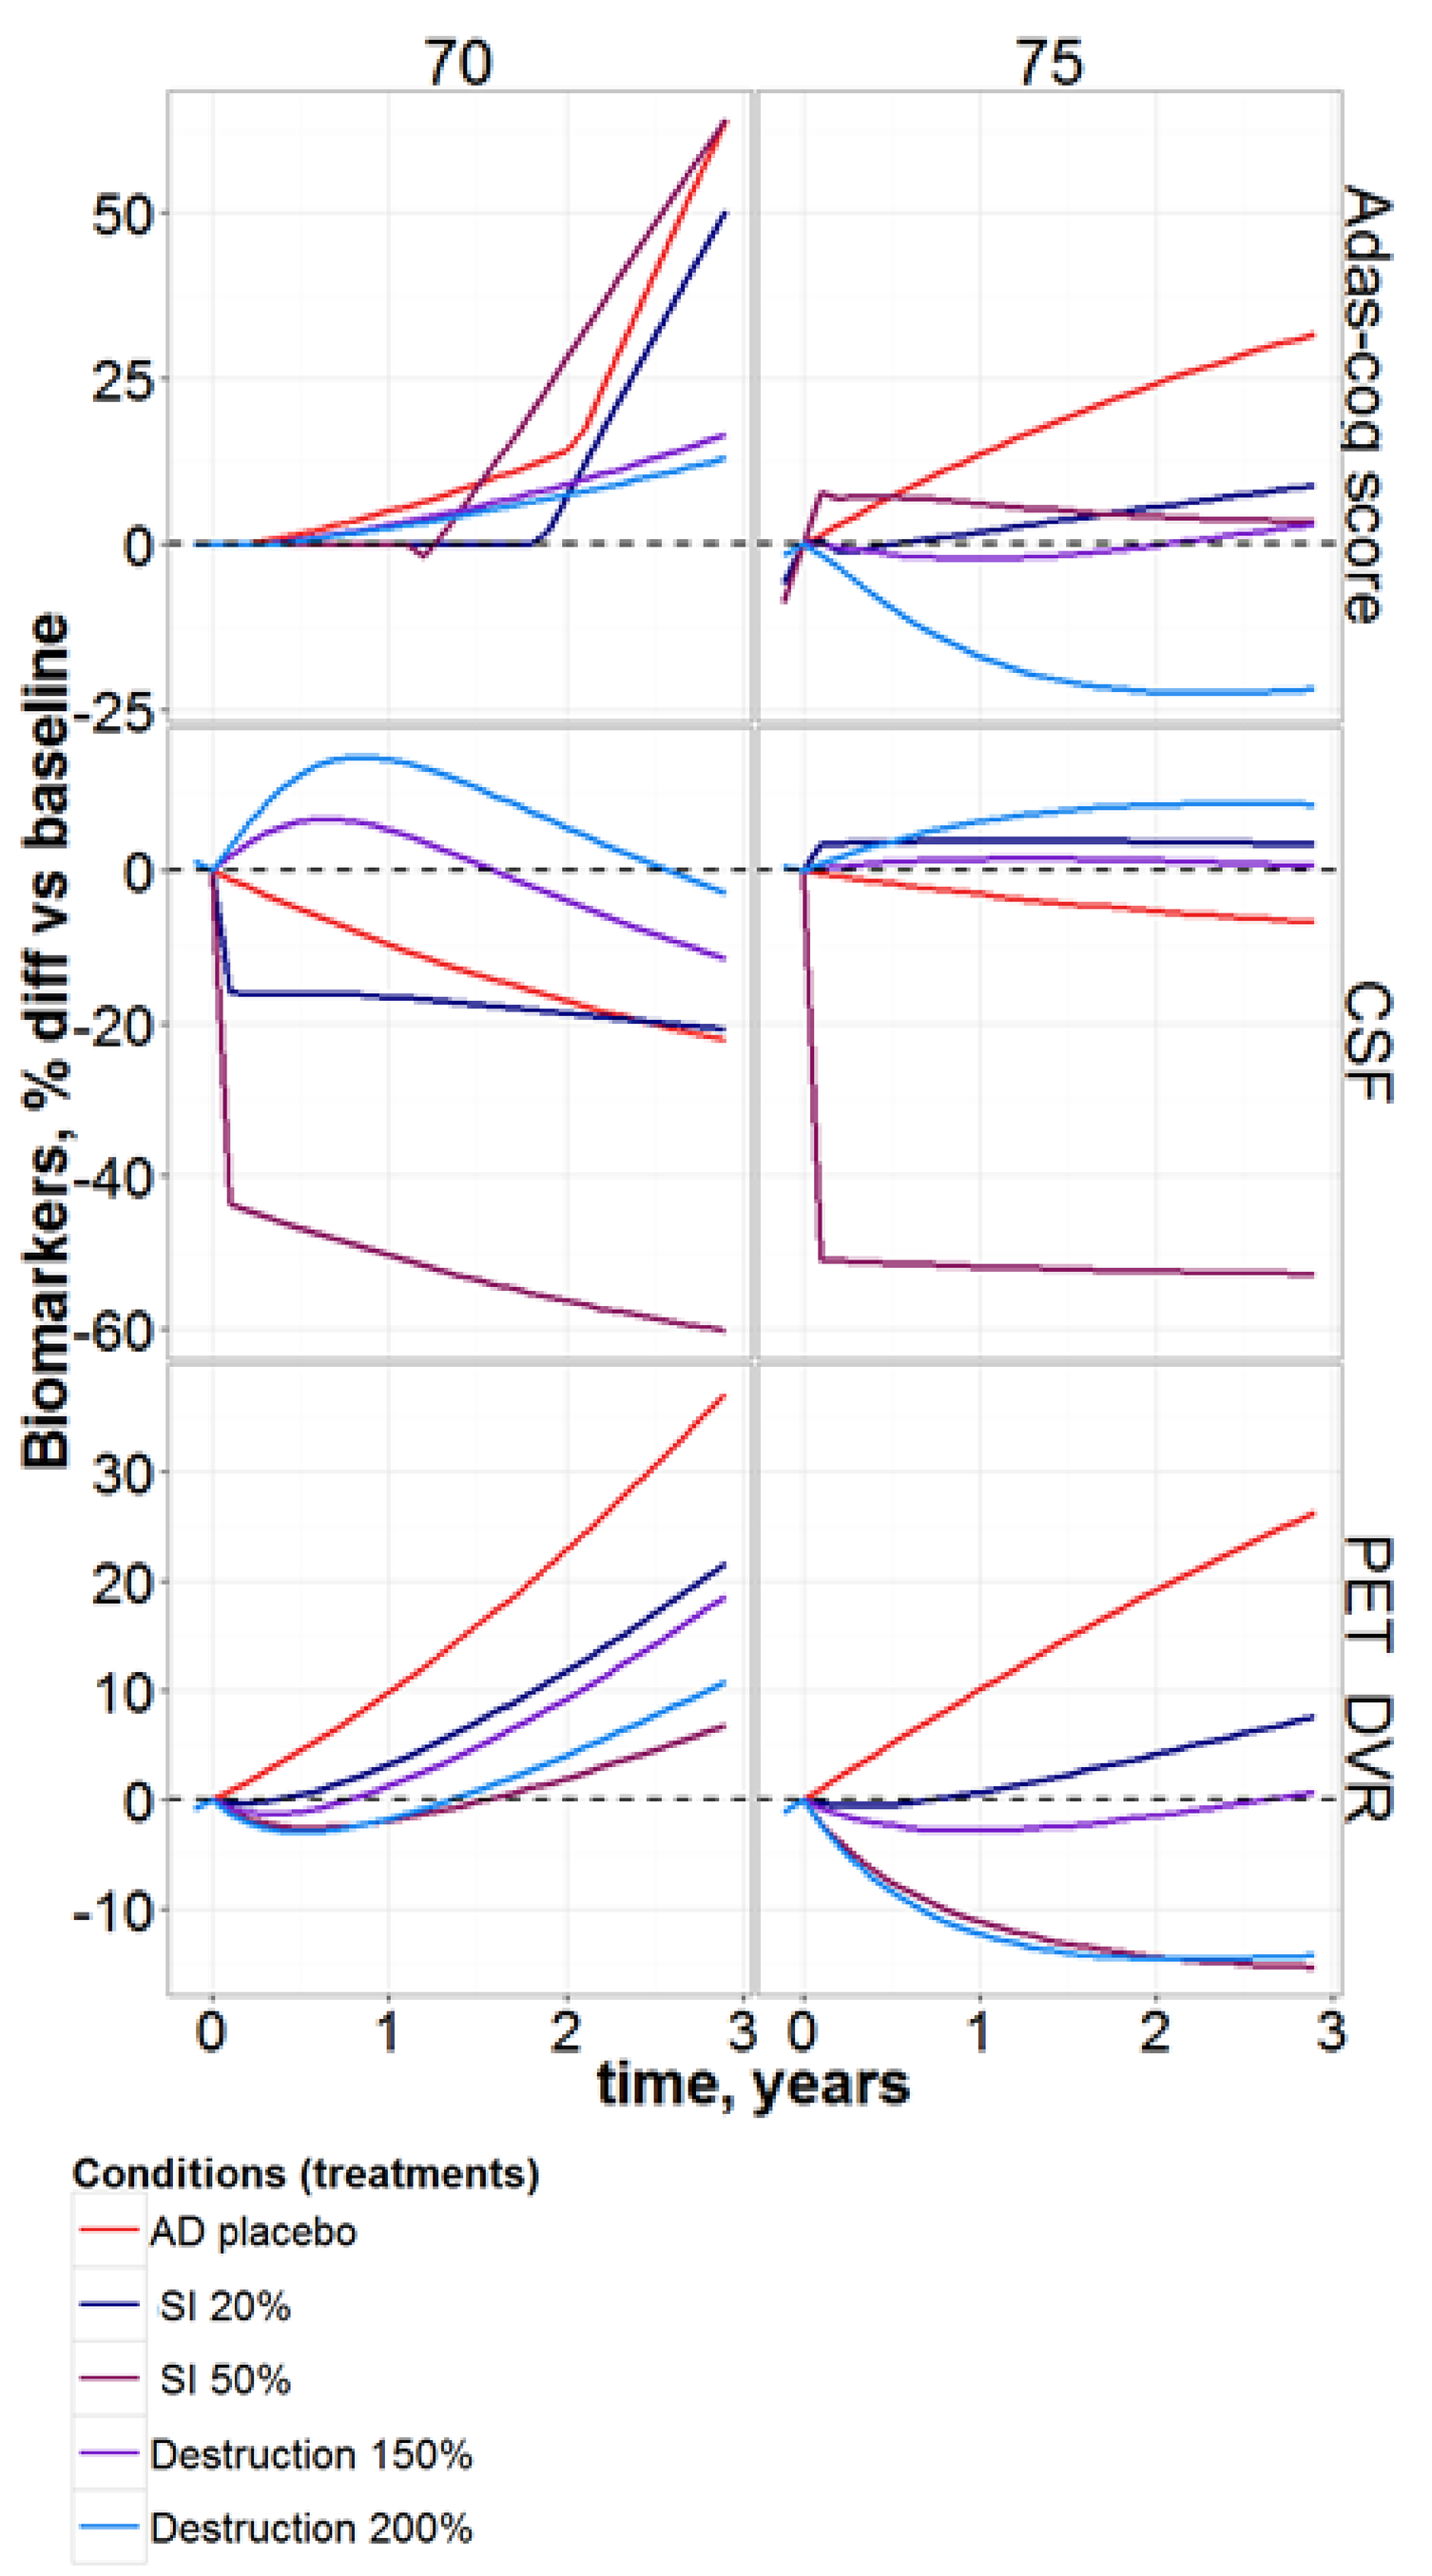
Age of start, years
70
75
Adas-cog score
CSF
PET DVR
Biomarkers, % diff vs baseline
time, years
Conditions (treatments)
AD placebo
SI 20%
SI 50%
Destruction 150%
Destruction 200%

Fig. 6 Validation of H 2.1 on Phase 2 trial data for **avagacestat** (Coric 2012).

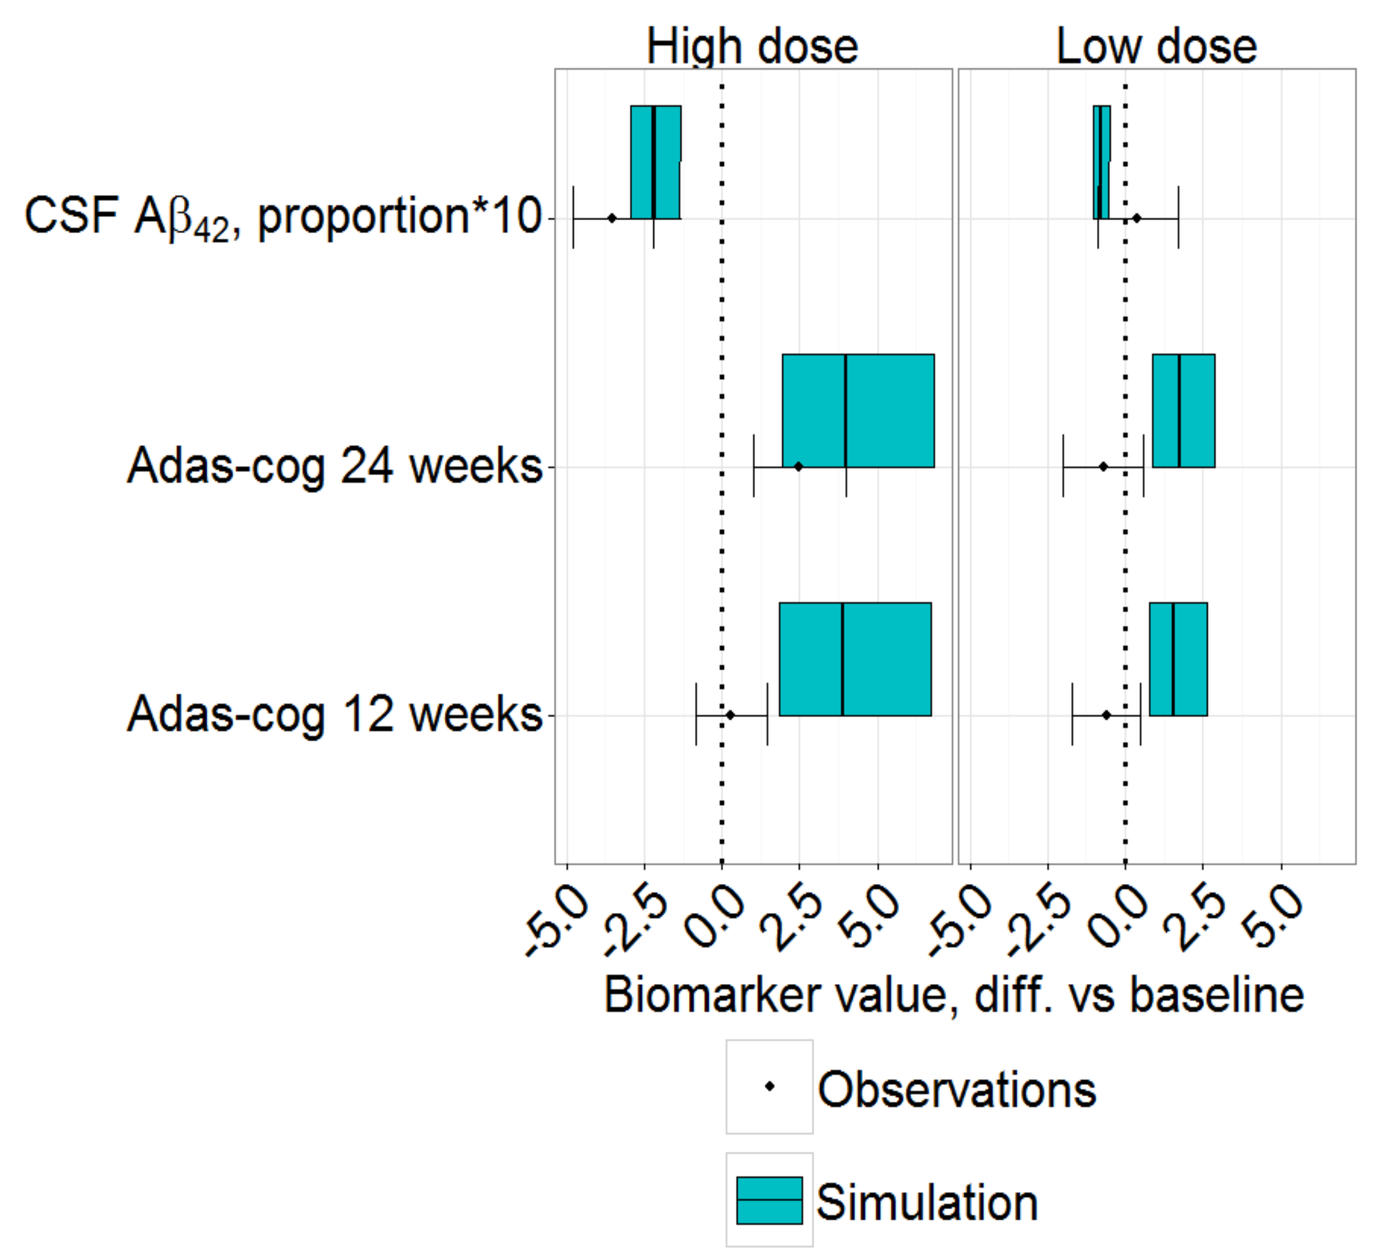
High dose
Low dose
CSF Aβ42, proportion*10
Adas-cog 24 weeks
Adas-cog 12 weeks
Biomarker value, diff. vs baseline
Observations
Simulation

Fig. 7. Validation of H 2.1 on Phase 2 data for **bapineuzumab** (Rinne 2010).

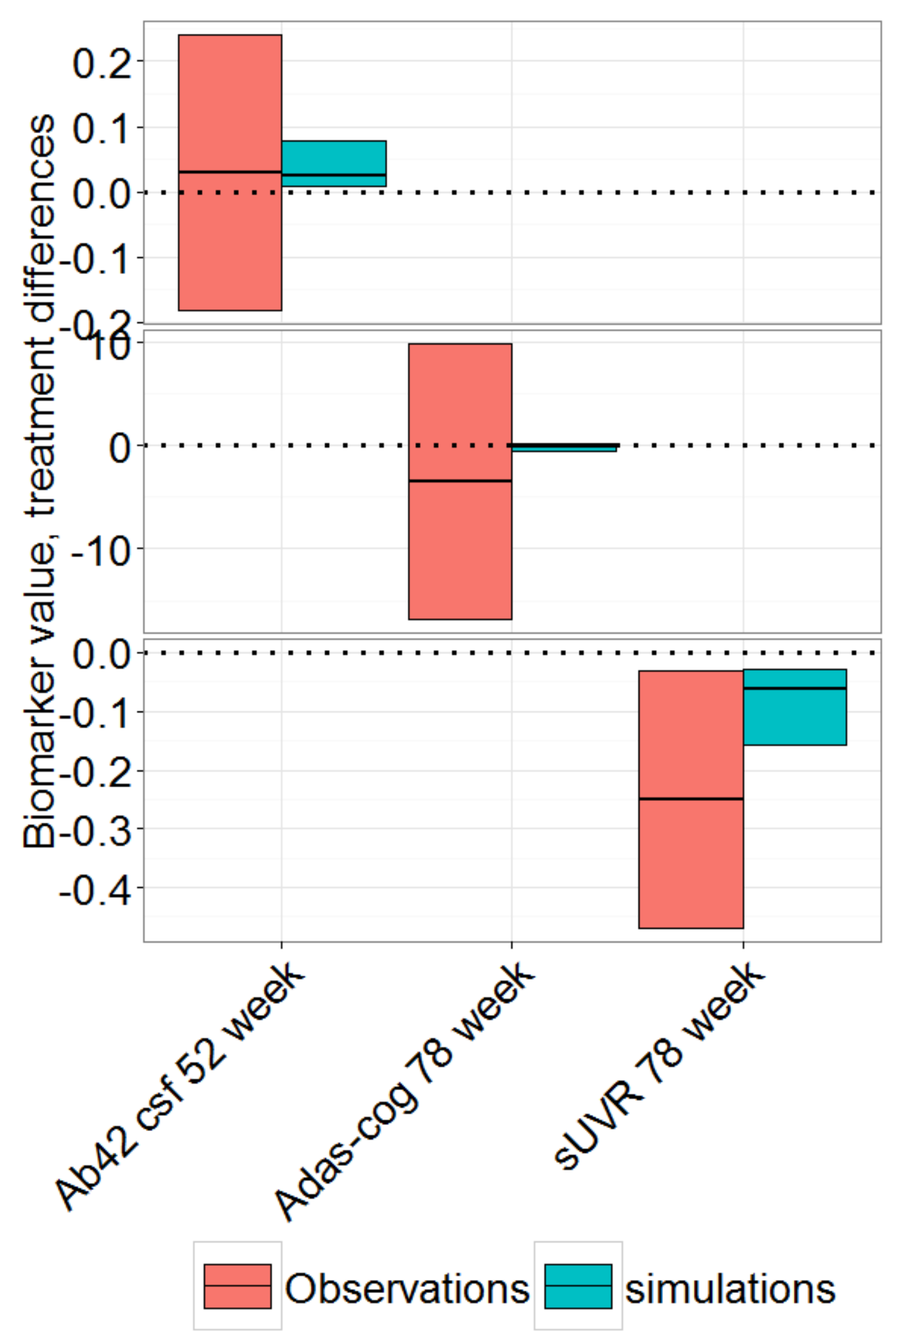
Biomarker value, treatment differences
Ab42 csf 52 week
Adas-cog 78 week
sUVR 78 week
Observations
simulations

CONCLUSIONS

- Only several designs may be used for legible confirming or rejection of the corresponding hypothesis
- For the threshold hypotheses no positive effect would be seen unless inhibition is high enough to overcome the toxic threshold (50 % inhibition)
- No negative effect predicted at destruction activation for any of the hypotheses
- Model predictions under hypothesis of proportional toxicity (and negative effect of high secretion inhibition) are successfully validated on CT results for two different therapies.

REFERENCES

1. Puzzo D, et al. J Neurosci. 2008 Dec 31;28(53):14537–45.
2. Rosenberg PB et al. Am J Geriatr Psychiatry. 2013;21(3):272–8.
3. Coric V et al. Arch Neurol. 2012;69(11):1430.
4. Rinne JO et al. Lancet Neurol. 2010 Apr;9(4):363–72.

CONTACTS

For more information, please visit our web site at www.insysbio.ru.
Tatiana Karelina by e-mail: karelina@insysbio.ru.